An Antioxidant Cocktail-containing Cosmetic Product (Liftactiv Cure) Prevents Air Pollution-induced Skin Hyperpigmentation: Results from the ex vivo Düsseldorf Pollution Skin Test

S Grether-Beck[1], A Valois[2], H Brenden[1], I Uthe[1], I Felsner[1], D Kerob[2], J Krutmann[1]

[1]IUF, Düsseldorf, Germany

[2]Laboratoires Vichy, Paris, France

Disclosure of Conflicts of Interest

- Support for these studies was provided by Laboratoires Vichy.
- A. Valois and D. Kerob are full-time employees of Laboratoires Vichy.
- S Grether-Beck, H Brenden, I Uthe, and I Felsner, and J Krutmann have no relationships to disclose.
- J Krutmann has worked as a consultant for Laboratoires Vichy.

Rationale and Objectives

- Exposure to traffic-related air pollutants is associated with facial pigment spots and wrinkles.
- In order to study the underlying mechanisms, we have developed a standardized, robust *ex vivo* human skin model which allows application of ambient relevant, toxicologically well characterized, traffic-related diesel exhaust particles (DEP) onto the surface of human skin[1].
- We previously demonstrated with this model that topical exposure of human skin to DEP at environmentally relevant, non-toxic concentrations increased skin pigmentation, which was mediated at least in part by oxidative stress.

The objective was to evaluate whether a topical formulation containing antioxidants (vitamin C, E, neohesperidine and maritime pine polyphenols (Liftactiv Cure [Vichy]; LC) was effective in protecting human skin against air pollution-induced skin pigmentation/ aging.

[1]*Grether-Beck S, et al. Ambient relevant diesel exhaust particles cause skin pigmentation ex vivo and in vivo in human skin: The Düsseldorf Pollution Patch Test. J Invest Dermatol 2018 138: Abstr. 1209, p 205, Supplement 1.*

Methods

Test product

- Formulation LC containing vitamin C (15%), vitamin E (0.5%), neohesperidine and maritime pine polyphenols, as well as Vichy volcanic mineralizing water.

Ex vivo skin culture

- Human skin was obtained from belly and mamma reduction surgery after informed consent.

In vitro Düsseldorf Pollution Skin Test

- LC and vehicle were applied at a dose of 2mg/cm^2 0.5 h prior to 6µg/cm^2 DEP standard reference material (SRM1650b).
- SRM1650b was applied twice for samples harvested after 6 days (double exposure) and three times for samples harvested after 9 days (triple exposure).

Analyses

- Colorimetry expressed as individual typology angle (ITAº)
- Photographs
- Gene expression by real-time PCR

Treatment	Harvest after 6 days	Harvest after 9 days
untreated	1-3	19-21
Product 1 (Vehicle)	4-6	22-24
Product 2 (Liftactiv Cure)	7-9	25-27
6µg/cm^2 SRM1650b	10-12	28-30
Product 1 + 6µg/cm^2 SRM1650b	13-15	31-33
Product 2 + 6µg/cm^2 SRM1650b	16-18	34-36

Results

Effect on DEP-induced skin hyperpigmentation

- At Day 1 (before exposure to DEP), no differences in skin color were observed within the intended treatment groups.
- After exposure to DEP, the effect LC-treated skin was significantly different from the effect of the vehicle ($p<0.05$).
- Compared to DEP alone, the percentage inhibition (calculated as 100% minus remaining skin color) with LC treatment was 47%, 34% and 47% after 3, 6 and 9 days, respectively.
- Vehicle had no inhibitory effect.

After 3 days

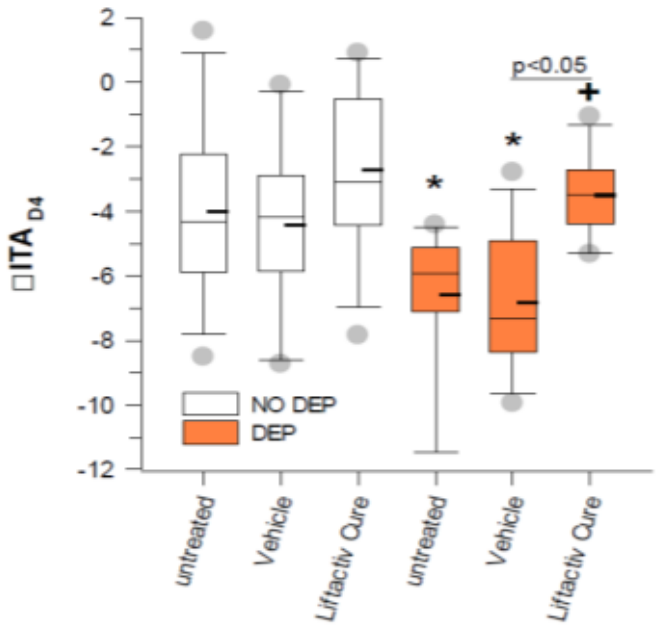

□ITA D4
2
0
-2
-4
-6
-8
-10
-12
p<0.05
NO DEP
DEP
untreated
Vehicle
Liftactiv Cure
untreated
Vehicle
Liftactiv Cure

After 6 days

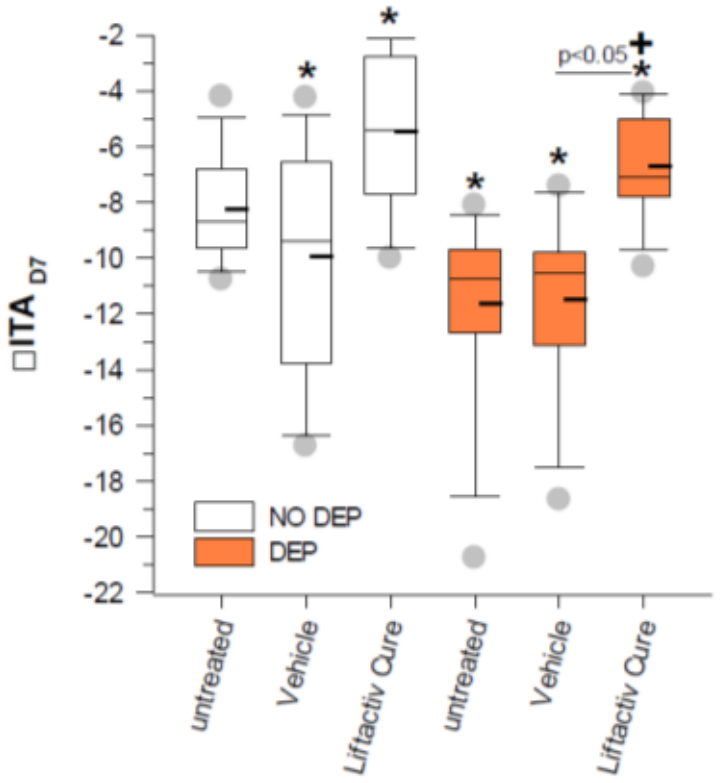

□ITA D7
-2
-4
-6
-8
-10
-12
-14
-16
-18
-20
-22
p<0.05
NO DEP
DEP
untreated
Vehicle
Liftactiv Cure
untreated
Vehicle
Liftactiv Cure

After 9 days

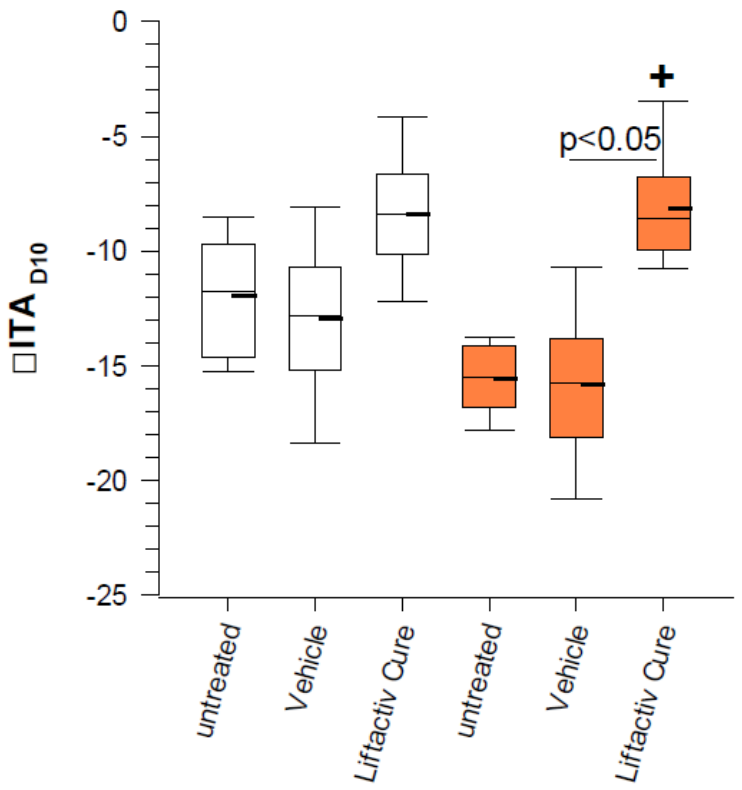

□ITA D10
0
-5
-10
-15
-20
-25
p<0.05
untreated
Vehicle
Liftactiv Cure
untreated
Vehicle
Liftactiv Cure

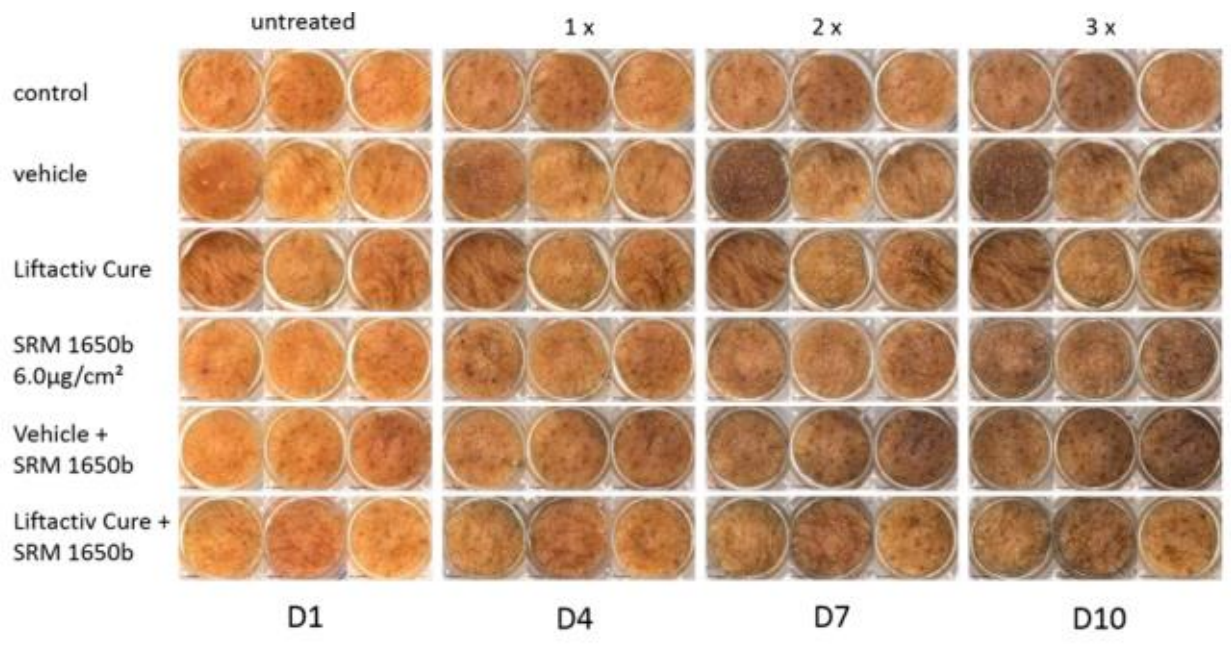

untreated
1 x
2 x
3 x
control
vehicle
Liftactiv Cure
SRM 1650b 6.0μg/cm²
Vehicle + SRM 1650b
Liftactiv Cure + SRM 1650b
D1
D4
D7
D10

Skin color changes visible in photographs

Application of LC significantly protected against the pollution-induced skin darkening effect, whereas the vehicle did not inhibit this DEP-induced pigmentation

Effect on DEP-induced Gene Expression

- Significant decrease of DEP-induced upregulation of IL1α, IL6, IL8, IL10, TNFα and PMEL with LC compared to vehicle at Day 7
- Significant decrease of IL1α, IL10, TNFα, and EDN with LC compared to vehicle at Day 10 (Table)

	% inhibition of DEP-induced upregulation	
DEP ±	Vehicle	Formulation LC
MMP1	24	32
IL1α	66	87
IL6	68	43
IL8	78	75
IL10	39	59
TNFα	61	90
CYP1A1	NA	22
POMC	NA	23
EDN	39	67
MITF	100	97
TYR	27	65
TRP	100	100
DCT	10	100
PMEL	83	33
MART	100	100

Formulation LC showed significant protection from pollution-induced expression of mainly proinflammatory cytokines

Active significantly better than vehicle
Vehicle significantly better than active

Conclusions

A formulation containing vitamin C, E, neohesperidine and maritime pine polyphenols as active ingredients significantly reduced DEP-induced skin pigmentation and gene expression of mainly proinflammatory cytokines. These studies emphasize that a formulation containing an appropriate cocktail of antioxidants is effective in protecting human skin against air pollution-induced skin pigmentation/ aging.